A phase 1 and pharmacokinetic study of ganetespib (STA-9090), a heat shock protein 90 inhibitor, in combination with docetaxel in patients with advanced solid tumor malignancies

R. D. Harvey[1,2], C. M. Lewis[2], J. S. Kauh[1,2], T. K. Owonikoko[1,2], A. Akintayo[2], M. Karol[3], F. Teofilovici[3], F. R. Khuri[1,2], S. S. Ramalingam[1,2]

[1]Department of Hematology and Medical Oncology, Emory University School of Medicine, Atlanta, GA, USA; [2]Winship Cancer Institute of Emory University, Atlanta, GA, USA; [3]Synta Pharmaceuticals Corp., Lexington, MA, USA

BACKGROUND

Hsp90 Inhibition

- Hsp90 is a chaperone protein that controls the folding and processing of proteins that drive tumor development and progression (**Figure 1**)
- Hsp90 clients include many proteins that play a critical role in tumor pathophysiology, such as EGFR, HER2, c-MET, ALK, AKT, BCR-ABL, RAF, CDK4, c-KIT, FLT3, and VEGFR[1]
- Degradation of client proteins allows for simultaneous targeting of multiple oncogenic signaling pathways
- Kinase client proteins are generally dependent on Hsp90 regardless of mutational status (ie, wild-type, TKI-sensitive, TKI-resistant) which creates potential for use in multiple settings

Figure 1. Hsp90 Chaperoning Cycle

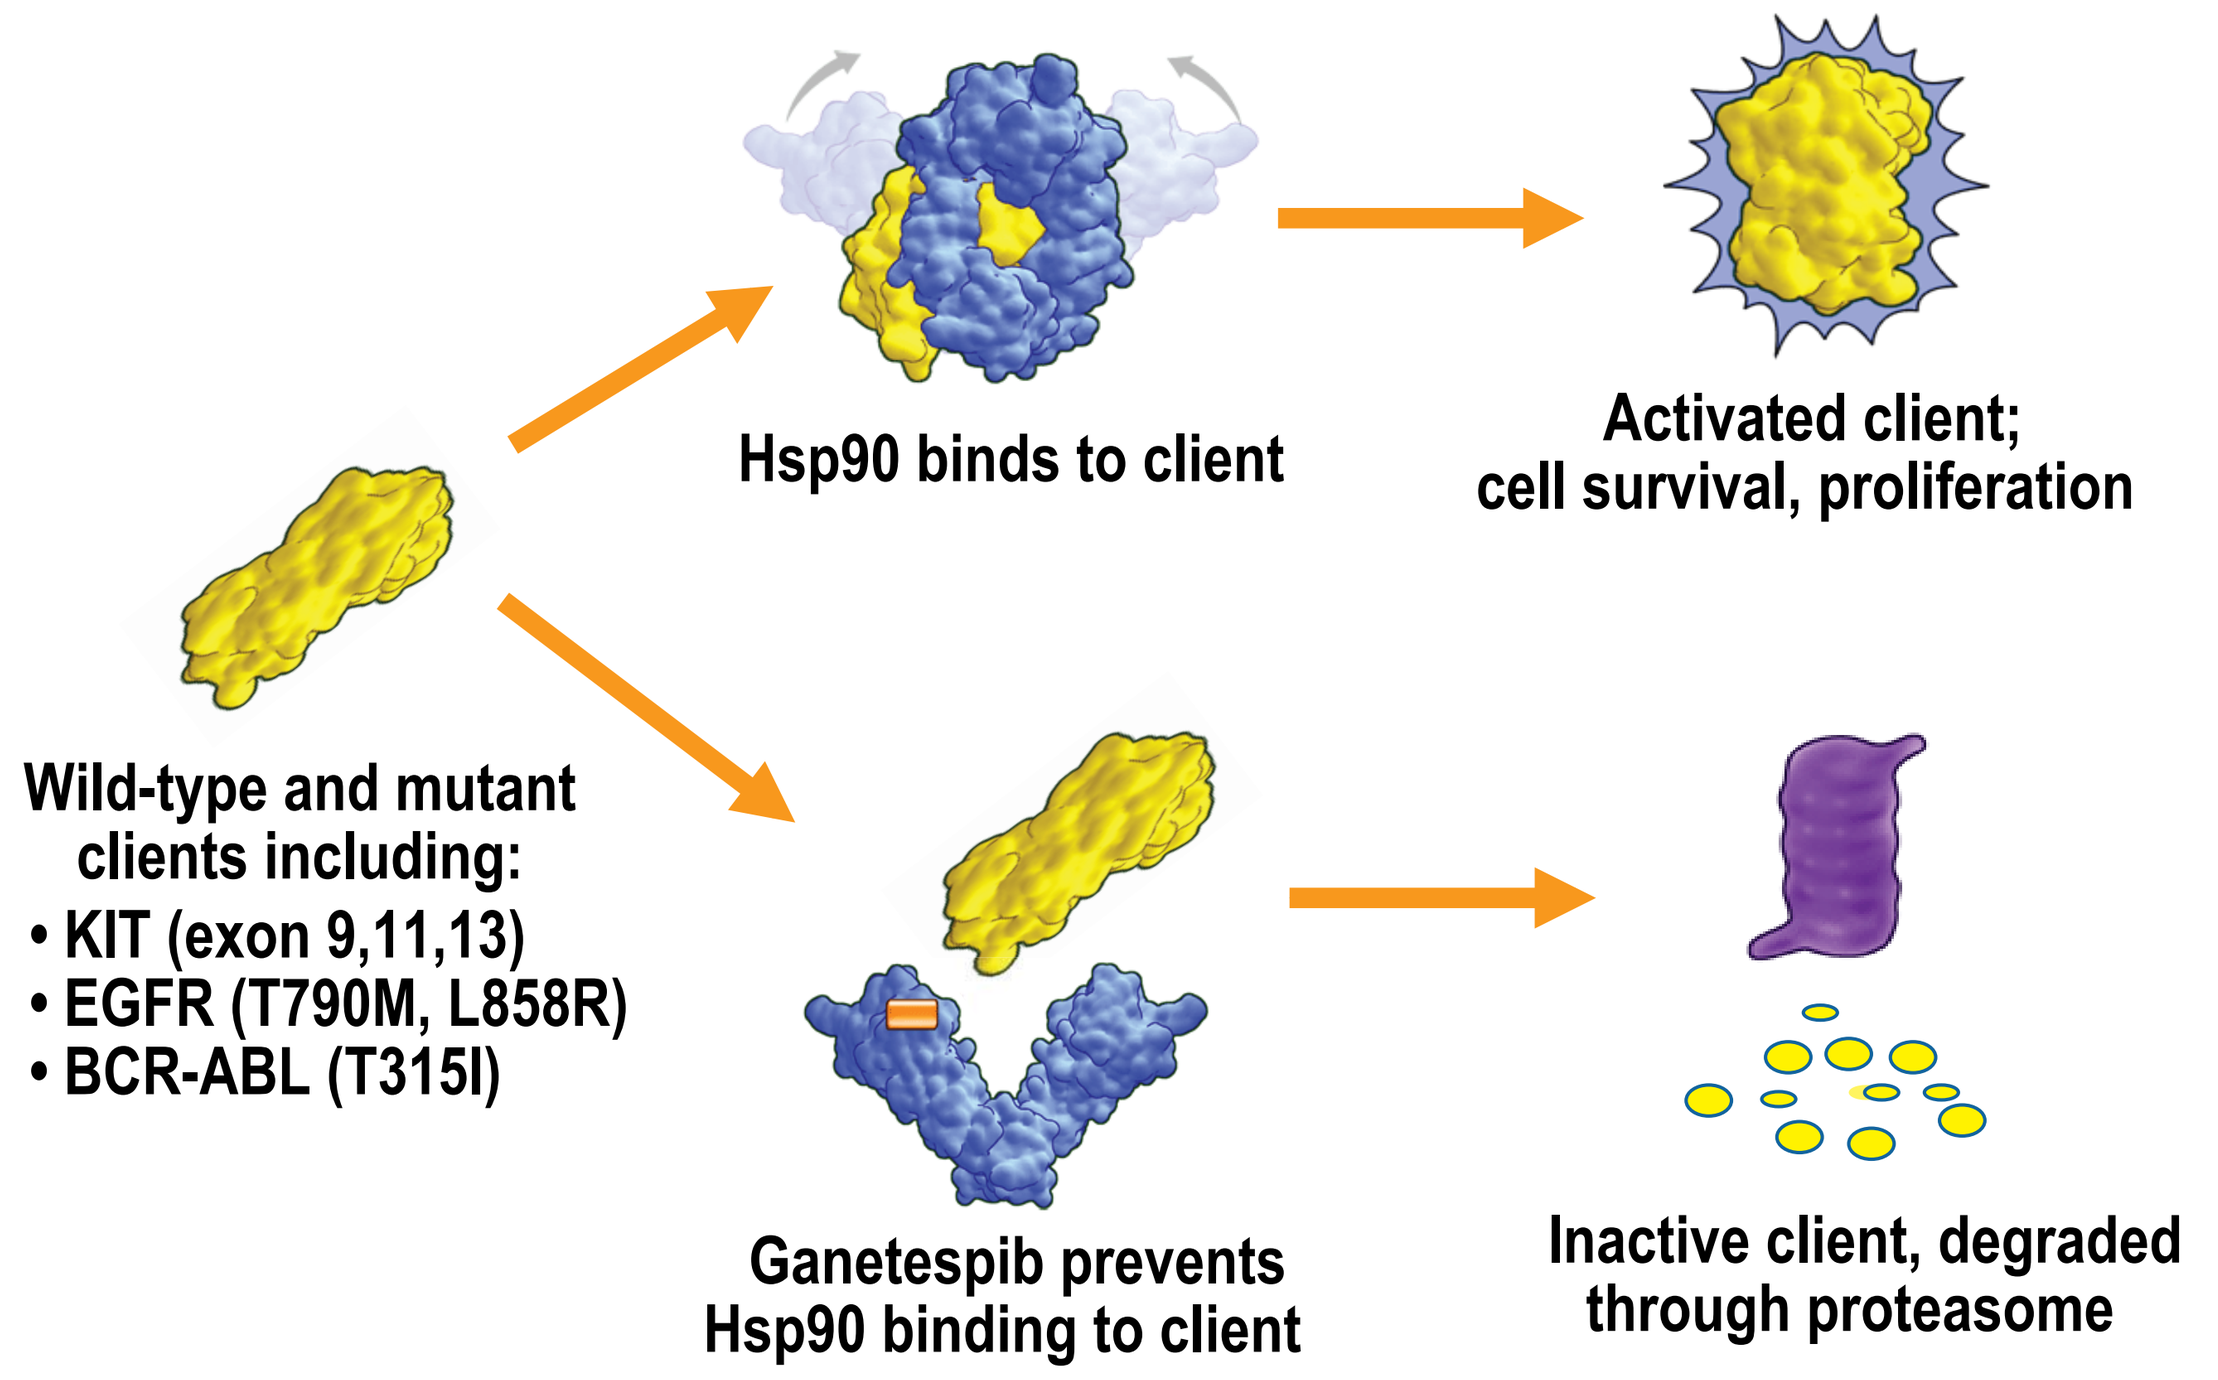
Hsp90 binds to client
Activated client; cell survival, proliferation
Wild-type and mutant clients including:
• KIT (exon 9,11,13)
• EGFR (T790M, L858R)
• BCR-ABL (T315I)
Ganetespib prevents Hsp90 binding to client
Inactive client, degraded through proteasome

Ganetespib

- Potent intravenous small molecule Hsp90 inhibitor[2-4]
 - Structurally unrelated to first-generation Hsp90 inhibitors (eg, 17AAG, IPI-504)
 - Up to 100 times more potent than first-generation Hsp90 inhibitors
- Translation of preclinical results
 - Potently inhibits Hsp90, leading to the degradation of multiple clinically validated oncogenic client proteins
 - Penetrates deeply into hypoxic tumors, inhibiting HIF-1α
- Clinically well-tolerated and has shown promising antitumor activity in early trials in multiple tumor types
- Shown to have synergy with docetaxel in preclinical studies

METHODS

Figure 2. Study Design

- Phase 1, open-label, dose-escalation
- Metastatic or unresectable solid tumors
- Disease progression prior to study entry
- ECOG PS 0-2
- Adequate organ function

Standard "3 + 3" design

Dose Escalation Phase (n=13)

Dose Level	Ganetespib (mg/m^2) Days 1 and 15	Docetaxel (mg/m^2) Day 1
1	150	60
2	150	75
3	200	75

Determine recommended dose

Expansion cohort for safety and tolerability (n=6)

ECOG, Eastern Cooperative Oncology Group; PS, performance status.

- 21-day cycles
- Ganetespib dosing changed from Days 1 and 8 to Days 1 and 15 following docetaxel-induced neutropenia observed on Day 8
- Each drug administered as a 1-hour infusion (1-hour rest in between)
- Pharmacokinetic sampling performed on Days 1 and 15 of Cycle 1
- Treatment until disease progression or unacceptable tolerability/toxicity
- Response assessment every 6 weeks

METHODS (CONT.)

Dose-limiting Toxicities Definition

- Non-hematologic toxicity:
 - Any Grade 3 or higher non-hematologic toxicity except nausea, vomiting, or alopecia
 - Diarrhea, nausea, or vomiting (≥ Grade 3) that lasts longer than 48 hours despite maximal medical therapy
 - Hypersensitivity-related toxicity ≥ Grade 3 with pre-medication
- Hematologic toxicity:
 - Grade 4 thrombocytopenia (<25,000/μL)
 - Grade 4 neutropenia (<500/μL) that persists for >7 days
 - Grade 3 or 4 febrile neutropenia (defined as temperature ≥101°F with a neutrophil count ≤1,000 cells/mL)

Study Objectives

- Primary Objective:
 - Determine the recommended doses for the combination of ganetespib and docetaxel in patients with advanced solid tumor malignancies
- Secondary Objectives:
 - Define the dose-limiting toxicities associated with the combination
 - Evaluate the possible effects of docetaxel on ganetespib pharmacokinetics, and vice versa
 - Quantify ganetespib and ganetespib metabolite exposures in the presence and absence of docetaxel
 - Characterize safety and tolerability of the combination
 - Assess preliminary evidence of antitumor activity

RESULTS

Table 1. Demographics and Baseline Characteristics

	Dose Level 1 (n=3)	Dose Level 2 (n=12)	Dose Level 3 (n=4)	Total (N=19)
Median age, y (range)	56 (44-63)	62 (51-73)	64 (63-70)	63 (44-73)
Gender, n (%)				
Male	1 (33)	6 (50)	0	7 (37)
Female	2 (67)	6 (50)	4 (100)	12 (63)
ECOG PS, n (%)				
0	0	2 (17)	0	2 (10)
1	3 (100)	10 (83)	4 (100)	17 (90)
Tumor type, n (%)				
NSCLC	1 (33)	3 (25)	2 (50)	6 (32)
Head and neck	0	3 (25)	0	3 (16)
Esophageal	1 (33)	0	0	1 (5)
Other	1 (33)	6 (50)	2 (50)	9 (47)
Prior systemic therapy, n (%)				
1	1 (33)	1 (8)	0	2 (10)
2-3	2 (67)	8 (67)	1 (25)	11 (58)
>3	0	3 (25)	3 (75)	6 (32)

ECOG, Eastern Cooperative Oncology Group; NSCLC, non–small-cell lung cancer; PS, performance status.

European Society of Medical Oncology at the 2011 European Multidisciplinary Cancer Congress

Stockholm, Sweden

Abstract Number 1.230

RESULTS (CONT.)

Treatment

- All 19 patients had at least 1 dose of study treatment
 - Median number of cycles: 4 (range, 1-11)
 - Median duration of treatment: 64 days (range, 1-234) (**Figure 3**)

Figure 3. Duration of Treatment

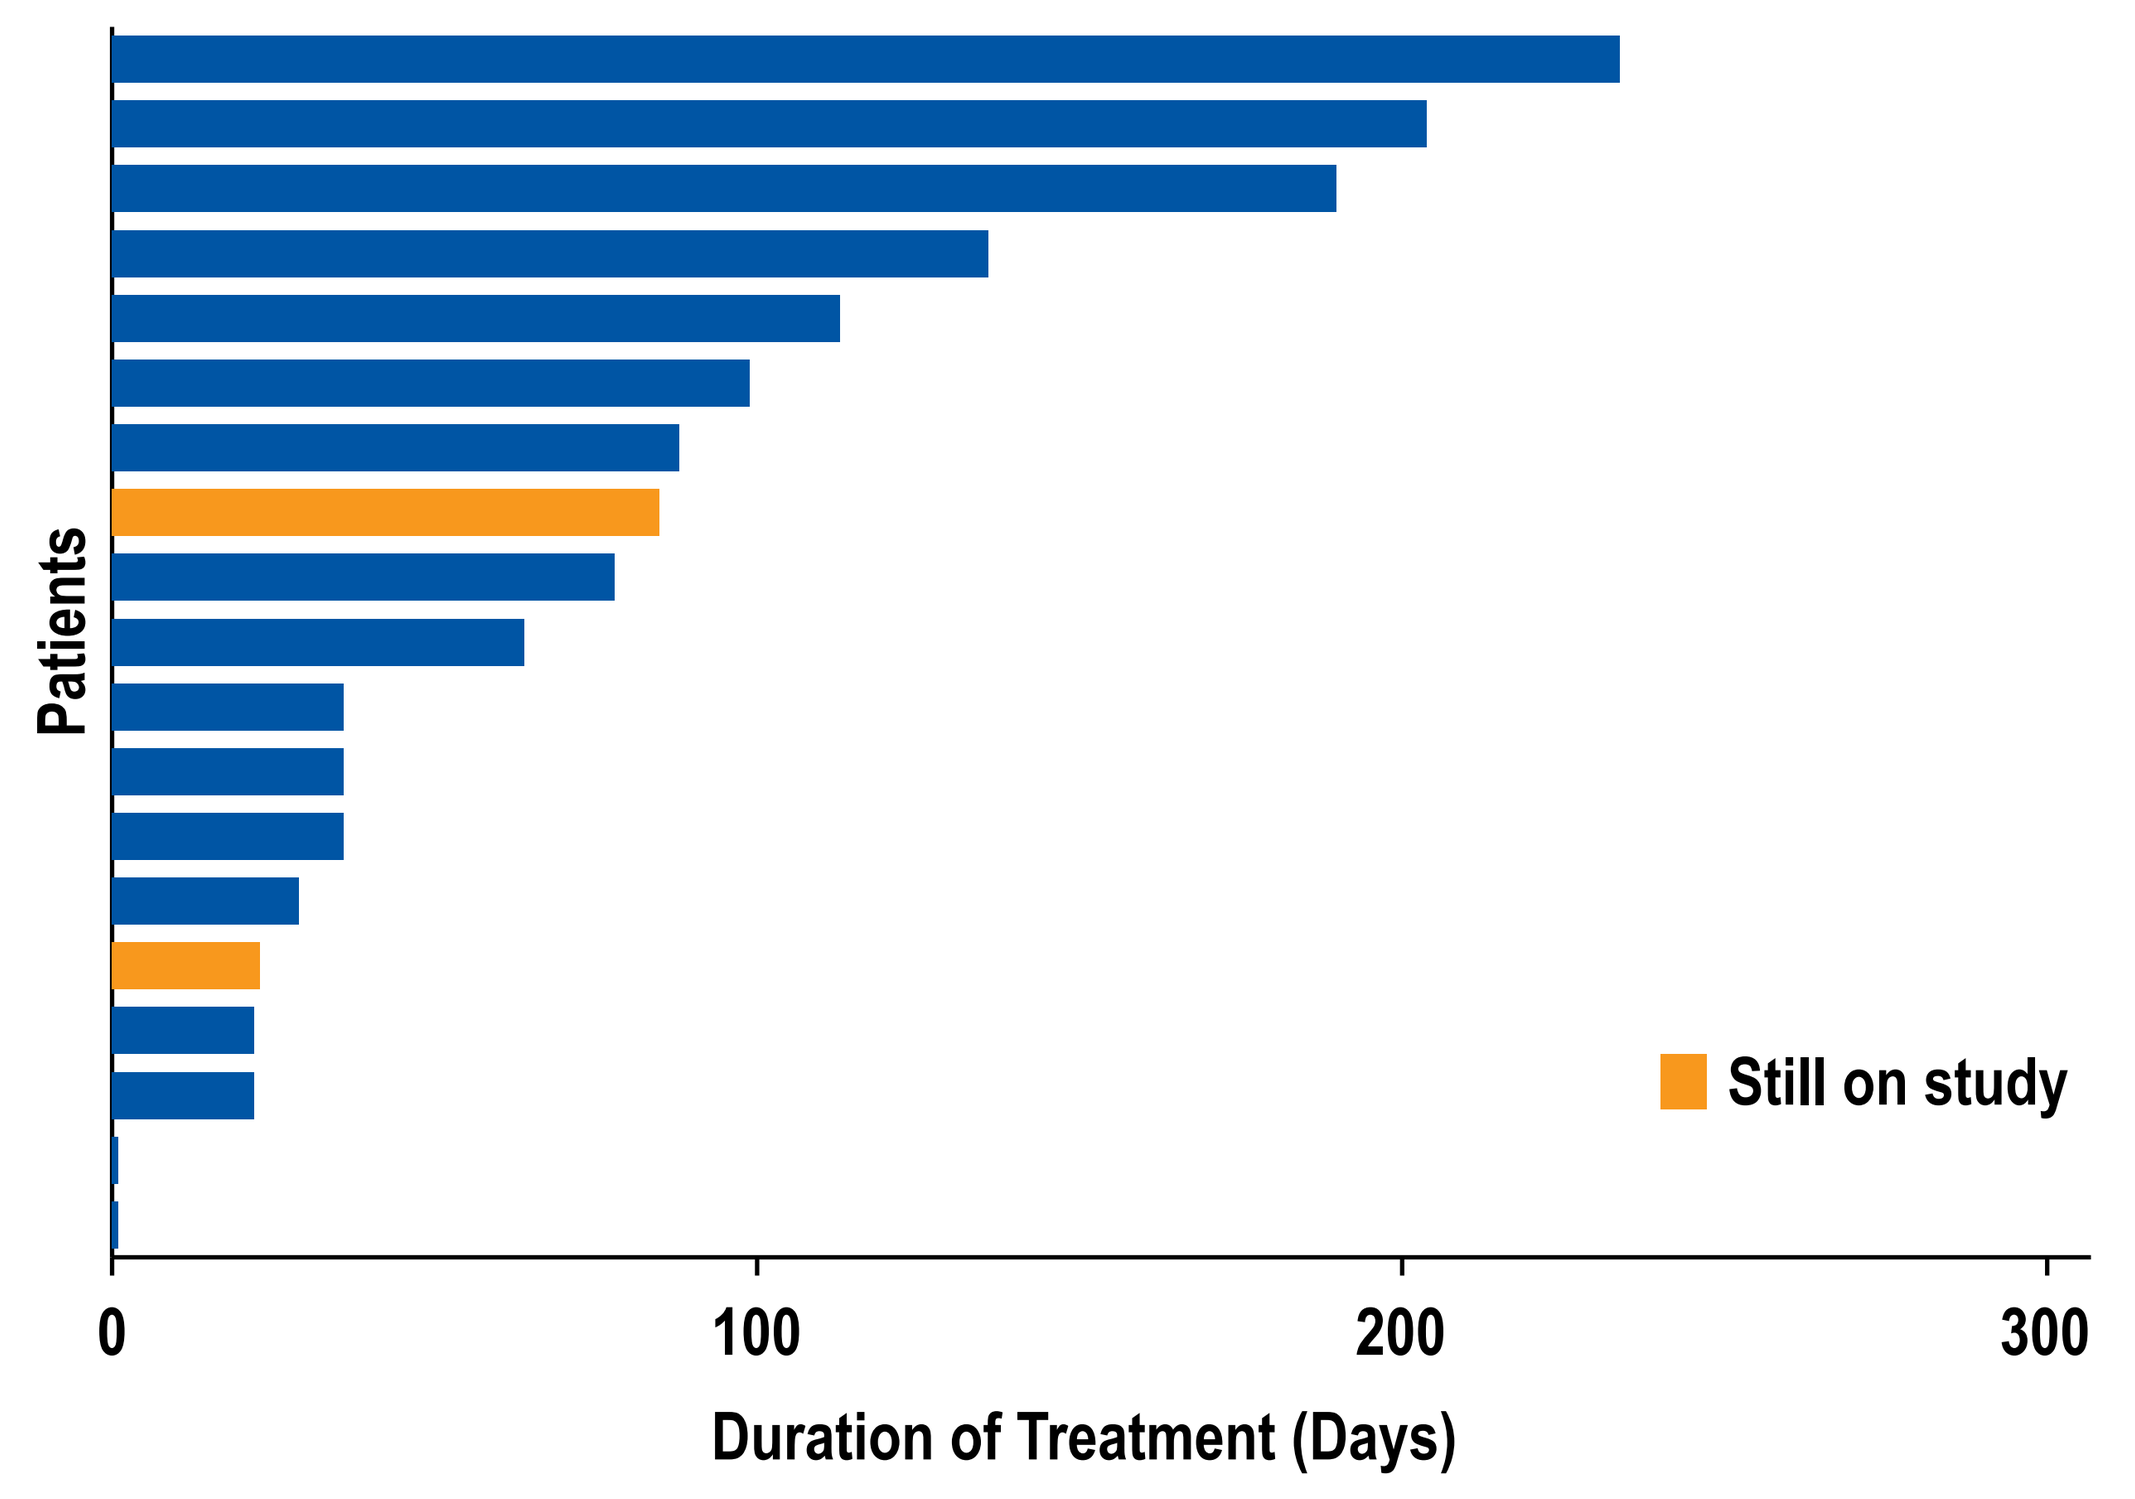
Patients
Still on study
0
100
200
300
Duration of Treatment (Days)

- 58% of patients had stable disease at the first assessment (end of Cycle 2)

Table 2. Summary of Treatment-emergent Adverse Events

Patients, n (%)	Dose Level 1 (n=3)	Dose Level 2 (n=11)	Dose Level 3 (n=4)	Total (N=18)
Any AE	3 (100)	11 (100)	4 (100)	18 (100)
Any AE ≥ Grade 3	3 (100)	9 (82)	4 (100)	16 (89)
Any SAE	1 (33)	7 (64)	1 (25)	9 (50)
Any DLT	0	1 (9)	2 (50)	3 (17)
Any AE leading to treatment discontinuation	1 (33)	1 (9)	1 (25)	3 (17)
Any AE with outcome of death	0	0	0	0

AE, adverse event; DLT, dose-limiting toxicity; SAE, serious adverse event.

- 3 AEs leading to discontinuation of study drug were serious adverse events (SAEs) of chest pain, mucosal inflammation, and febrile neutropenia

Figure 4. Treatment-emergent Adverse Events (≥10% of Patients)

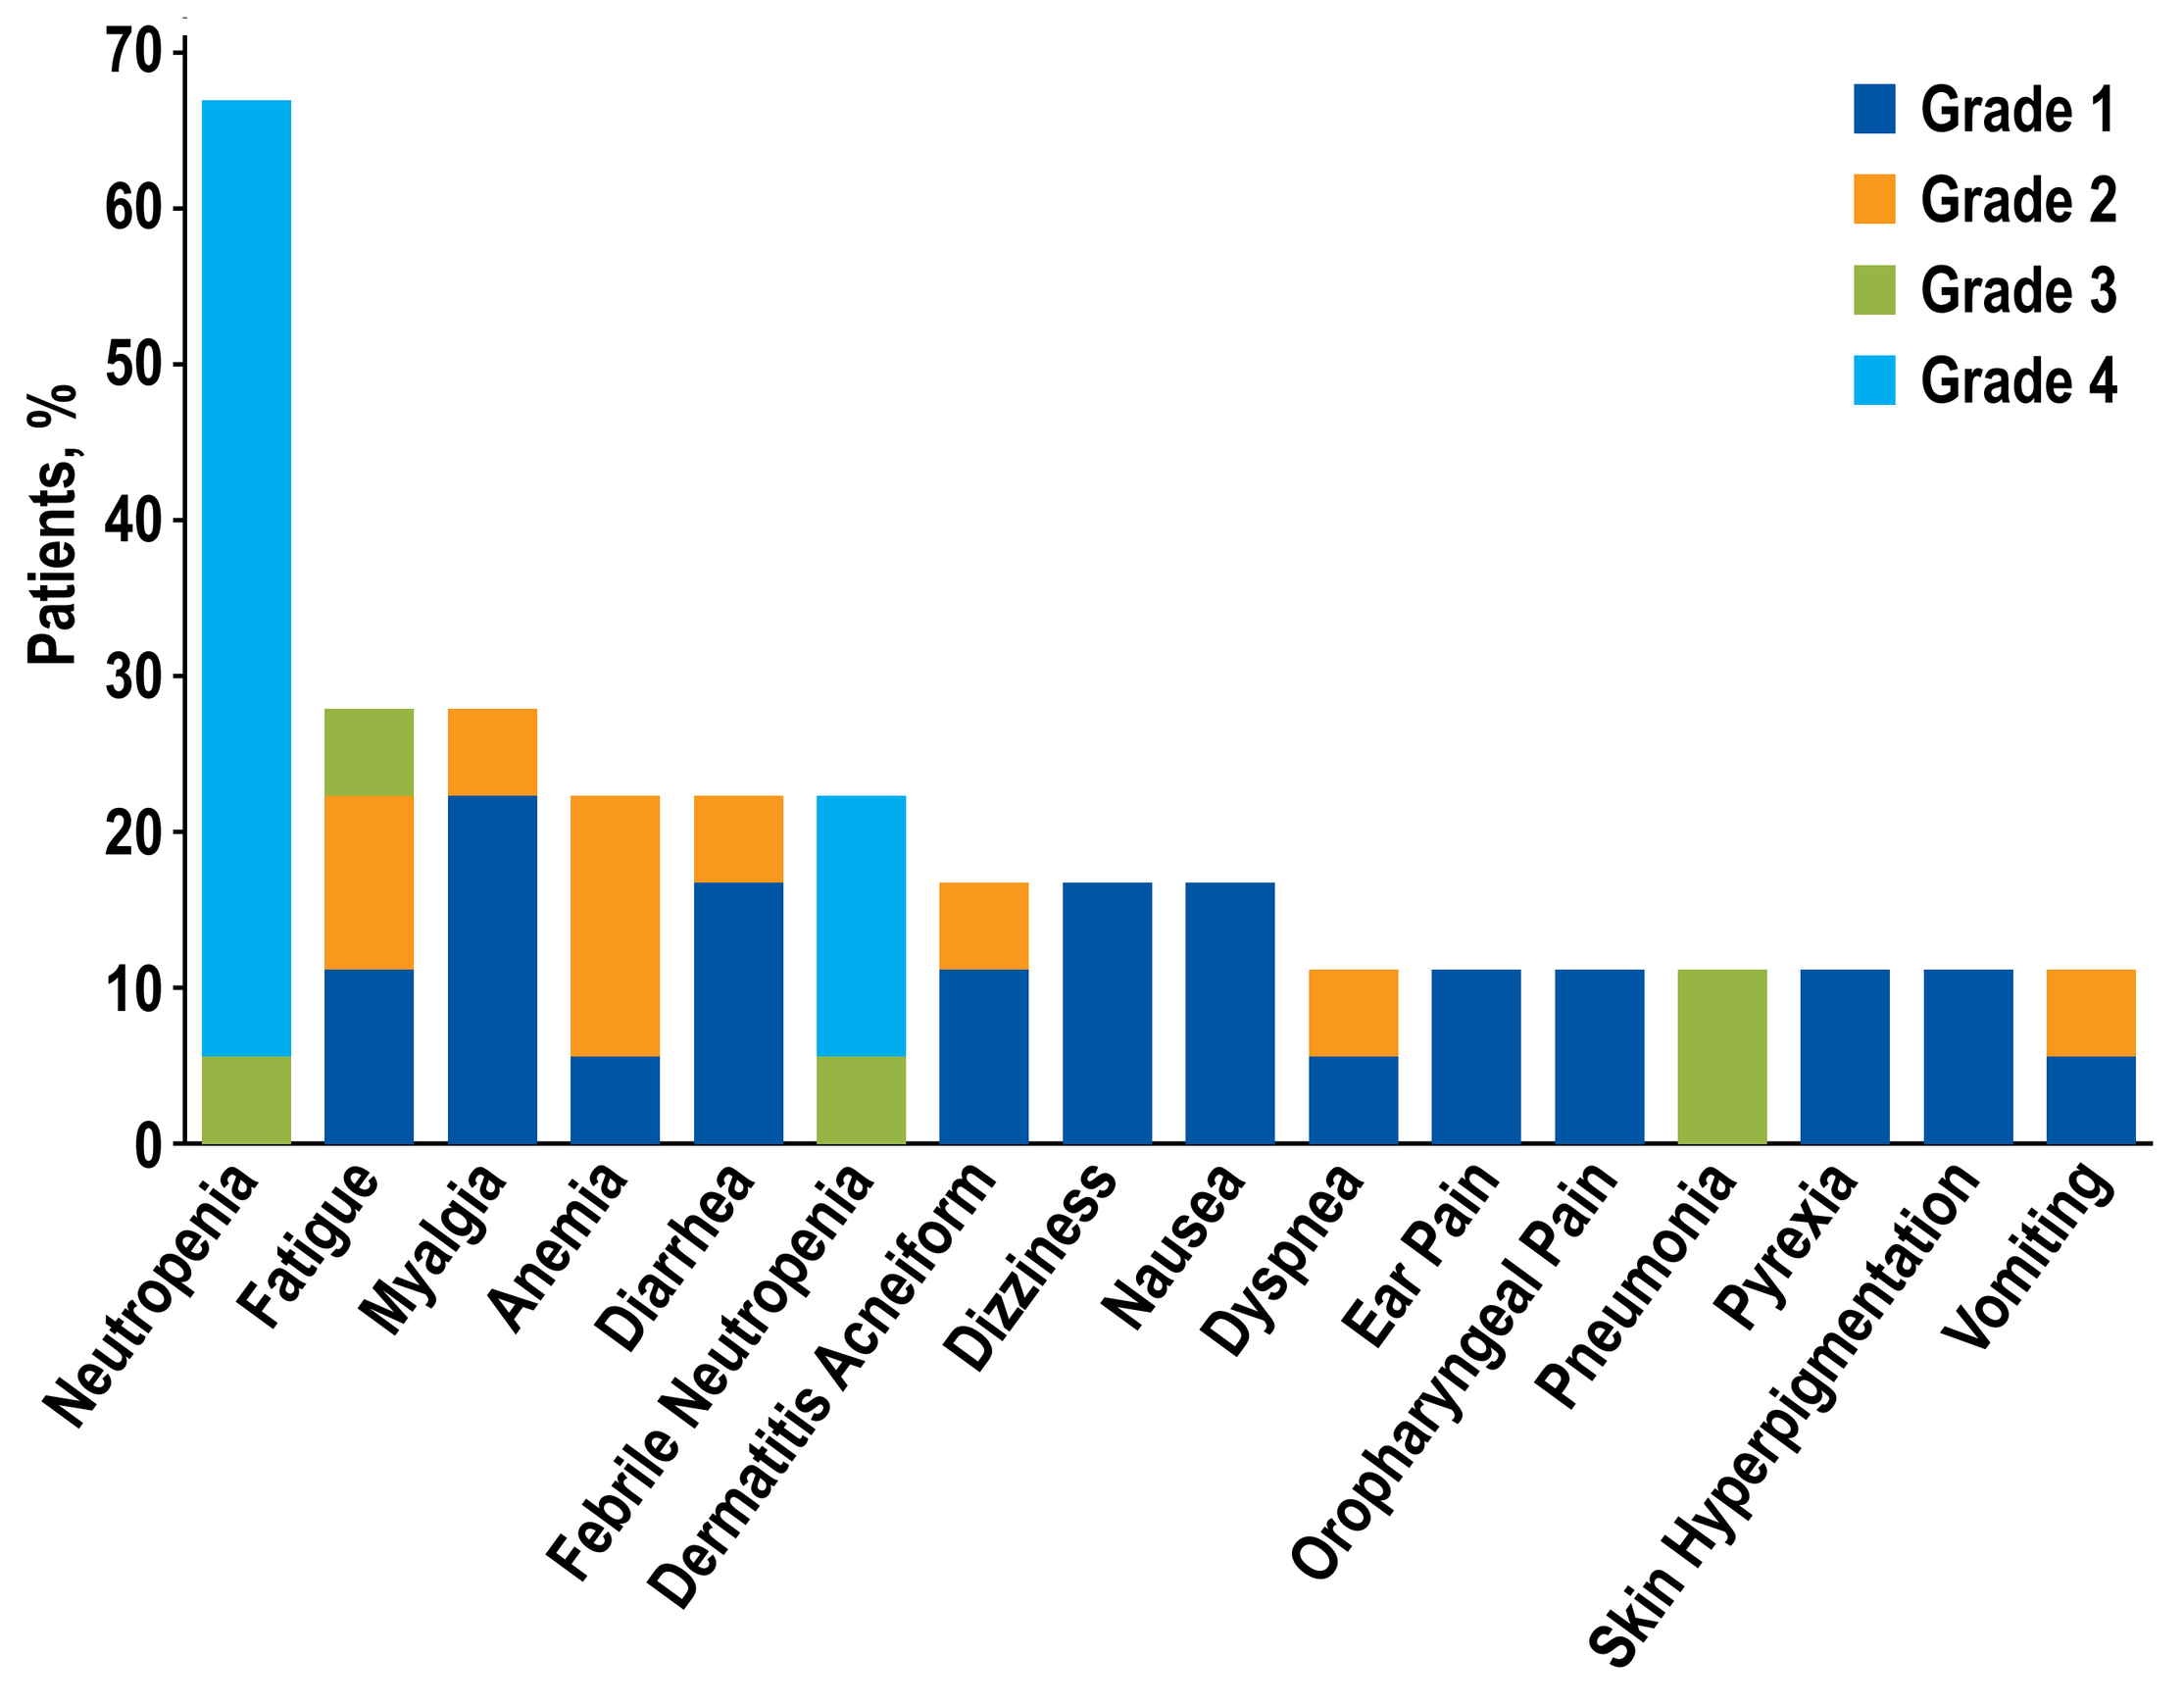
Patients, %
0
10
20
30
40
50
60
70
Grade 1
Grade 2
Grade 3
Grade 4
Neutropenia
Fatigue
Myalgia
Anemia
Diarrhea
Febrile Neutropenia
Dermatitis Acneiform
Dizziness
Nausea
Dyspnea
Ear Pain
Oropharyngeal Pain
Pneumonia
Pyrexia
Skin Hyperpigmentation
Vomiting

- Docetaxel-induced neutropenia was common approximately 8 days following dosing
 - Typically resolved within 7 days

Dose-Limiting Toxicities

- Dose level 1 (ganetespib 150 mg/m^2, docetaxel 60 mg/m^2): 0/3 patients
- Dose level 2 (ganetespib 150 mg/m^2, docetaxel 75 mg/m^2): 1/6 patients
 - Mucosal inflammation (Grade 3, SAE, resolved following drug discontinuation)
- Dose level 3 (ganetespib 200 mg/m^2, docetaxel 75 mg/m^2): 2/4 patients
 - Febrile neutropenia (Grade 4, SAE, resolved following drug discontinuation)
 - Neutropenia (Grade 4, resolved after 2 weeks)
- Maximum tolerated dose = ganetespib 150 mg/m^2 with 75 mg/m^2 docetaxel
- 6 additional patients enrolled into dose level 2

Serious Adverse Events

- SAEs were reported in 9 patients (50%)
- SAEs reported in more than 1 patient were febrile neutropenia (4 patients, 22%) and pneumonia (2 patients, 11%)
- SAEs reported in 1 patient each (6%) were chest pain, chills, dyspnea, fatigue, mucosal inflammation, neutropenia, pneumothorax, pulmonary embolism, rib fracture, transient ischaemic attack, and vomiting

Pharmacokinetics

- Ganetespib pharmacokinetics were consistent over time (Day 1 vs Day 15, **Table 3**, **Figure 5**)
- Ganetespib pharmacokinetics were linear as demonstrated by dose-independent T_{max}, $t_{1/2}$, V_z, and CL (150 mg/m^2 and 200 mg/m^2 doses, **Table 3**)
- Ganetespib pharmacokinetics were unaffected by docetaxel administration (Day 1 with docetaxel vs Day 15 without docetaxel, **Table 3**, **Figure 5**)
- Clinically, ganetespib plasma concentrations rose rapidly during infusion, achieved maximum rapidly and then declined rapidly to approximately 1/10 of C_{max} within the first hour and to approximately 1/100 of C_{max} at 10 hours following infusion termination (**Figure 5**)
- No effect of ganetespib on docetaxel pharmacokinetics (**Table 3**)

Table 3. Ganetespib and Docetaxel Pharmacokinetics

	T_{max} (h)		C_{max} (ng/mL)		$t_{1/2}$ (h)		V_z (L/m^2)		CL ($L/h/m^2$)	
	Day 1	Day 15	Day 1	Day 15	Day 1	Day 15	Day 1	Day 15	Day 1	Day 15
Ganetespib[a] 150 mg/m^2										
N	8	8	8	8	8	8	8	8	8	8
Mean	0.68	0.88	4963	4709	11.0	11.6	366.3	383.3	22.8	22.0
SD	0.25	0.25	1669	1561	1.5	2.0	125.7	204.3	5.5	7.1
CV%	36.4	27.9	33.6	33.1	13.4	17.5	34.3	53.3	24.3	32.0
Ganetespib[a] 200 mg/m^2										
N	4	3	4	3	4	3	4	3	4	3
Mean	0.73	0.66	7548	5197	11.1	11.8	270.4	476.6	16.8	29.5
SD	0.27	0.27	2205	2946	3.3	3.1	108.1	169.3	2.7	11.8
CV%	36.7	41.1	29.2	56.7	29.5	26.0	40.0	35.5	16.1	40.1
Docetaxel[b] 60 mg/m^2										
N	3		3		3		3		3	
Mean	0.50		2100		19.4		639.9		23.8	
SD	0.00		626		4.4		84.1		7.32	
CV%	0.0		29.8		22.6		13.1		30.8	
Docetaxel[b] 75 mg/m^2										
N	9		9		9		9		9	
Mean	0.75		2864		22.9		665.8		20.2	
SD	0.24		733		5.6		240.4		6.6	
CV%	31.9		25.6		24.5		36.1		32.5	

C_{max}, peak concentration; CL, clearance rate; CV, coefficient of variation; SD, standard deviation; T_{max}, time of peak concentration; $t_{1/2}$, half-life; V_z, apparent volume of distribution at terminal phase.
[a]Ganetespib sampling: 0, 0.5, 1, 2, 3, 4, 7, 9.5, 27, and 51 hours relative to infusion start.
[b]Docetaxel sampling: 0, 0.5, 1, 2, 5, 7.5, 25, and 49 hours relative to infusion start.

- Published docetaxel pharmacokinetics
 - Docetaxel mean clearance[5-7] ranges from 19.6 to 29.1 $L/h/m^2$
 - Docetaxel mean half-life[5,7] ranges from 9.6 to 18.5 hours

Figure 5. Ganetespib Profile With (Day 1) and Without (Day 15) Docetaxel

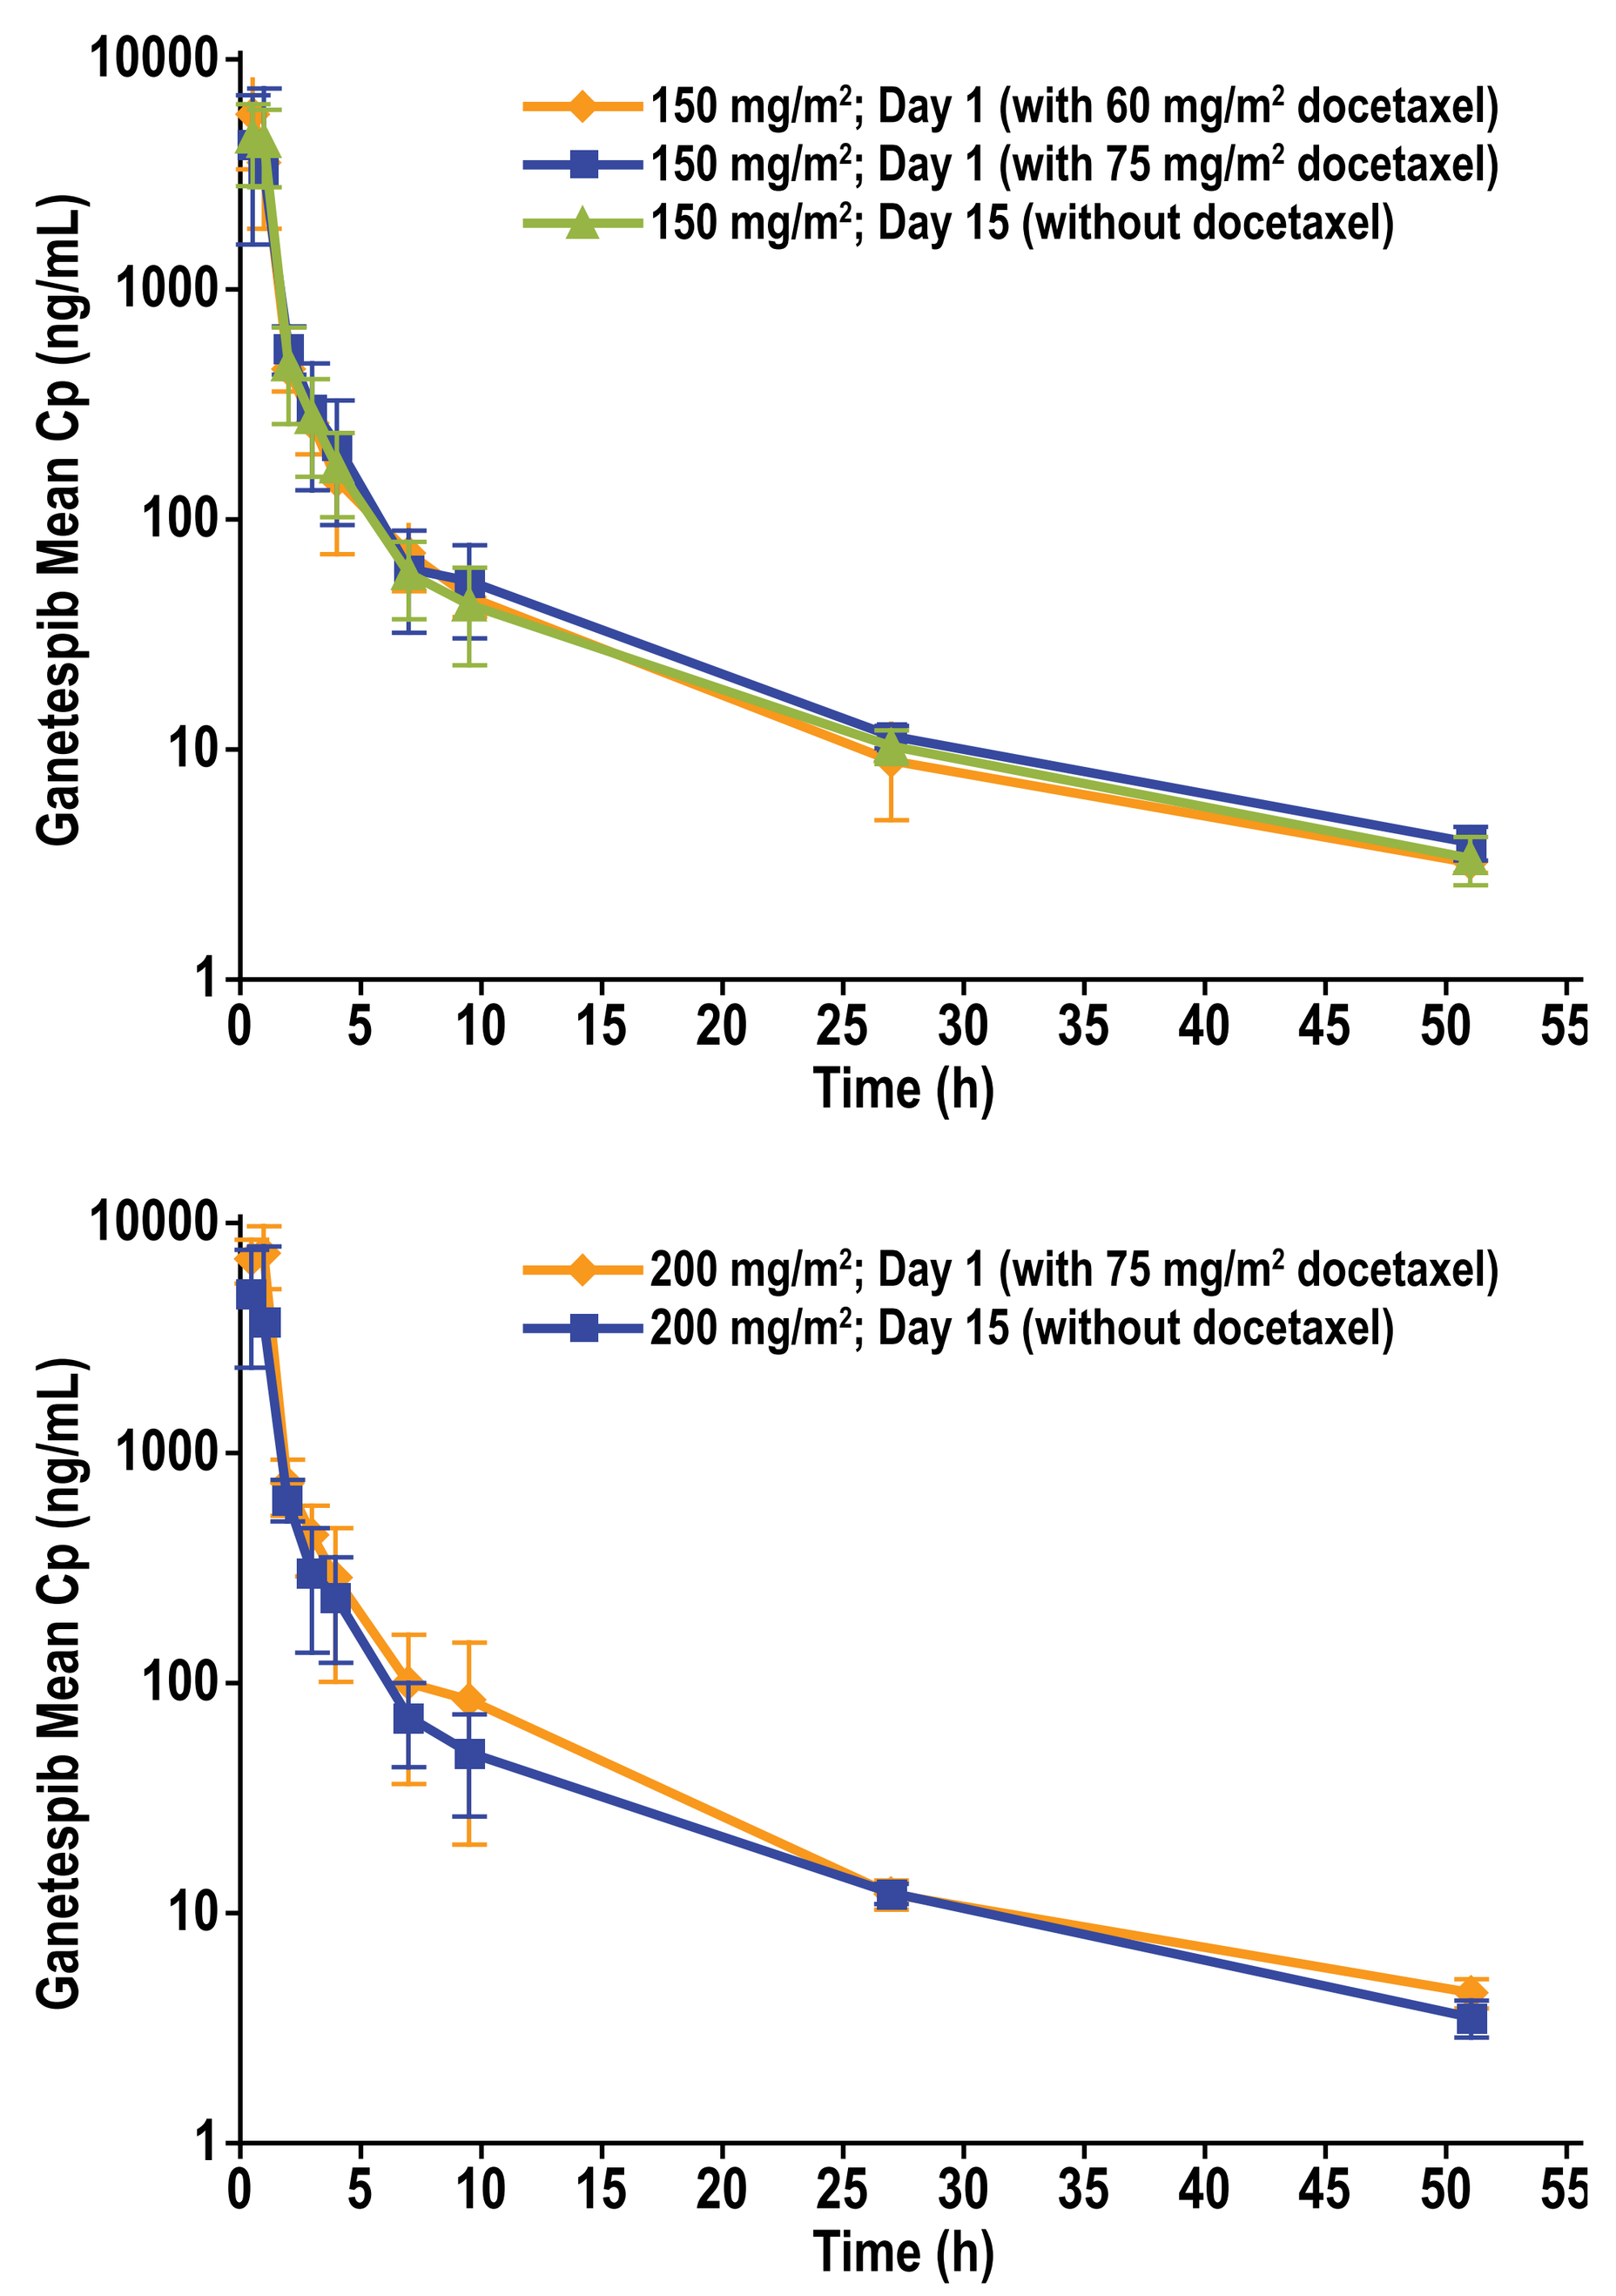
150 mg/m²; Day 1 (with 60 mg/m² docetaxel)
150 mg/m²; Day 1 (with 75 mg/m² docetaxel)
150 mg/m²; Day 15 (without docetaxel)
200 mg/m²; Day 1 (with 75 mg/m² docetaxel)
200 mg/m²; Day 15 (without docetaxel)
Ganetespib Mean Cp (ng/mL)
1
10
100
1000
10000
0
5
10
15
20
25
30
35
40
45
50
55
Time (h)

Cp, plasma concentration.

- No effect of docetaxel on ganetespib exposure

CONCLUSIONS

- The combination of ganetespib and docetaxel is well-tolerated with half of the patients receiving at least 4 cycles of treatment
- Neutropenia was common approximately 8 days post-docetaxel
 - Typically resolved spontaneously within 7 days
 - Consistent with known effects of docetaxel treatment[8-10]
- Pharmacokinetic data indicate pharmacokinetic similarity between ganetespib administered alone and ganetespib administered prior to docetaxel
- No drug accumulation observed following once-weekly dosing which is consistent with other studies of ganetespib monotherapy
- The recommended combination dose for future studies is 150 mg/m^2 ganetespib on Days 1 and 15 and 75 mg/m^2 docetaxel on Day 1 in a 21-day cycle
- A randomized phase 2b/3 study (NCT01348126) of the combination has been initiated in advanced non–small-cell lung cancer

Acknowledgements

The authors would like to thank the study participants and their families, as well as all of the study investigators and study team members. Editorial support in assembling tables and figures, collating author comments, copyediting, fact-checking, referencing, and graphic services were provided by AOI Communications, L.P.

Disclosures

R. D. Harvey, C. M. Lewis, J. S. Kauh, A. Akintayo, M. Karol, F. Teofilovici, F. R. Khuri, and S. S. Ramalingam have nothing to disclose. T. K. Owonikoko served on an advisory panel for Sanofi-Aventis.

References

1. Li Y, Zhang T, Schwartz SJ, and Sun D. *Drug Resist Updat.* 2009;12:17-27; **2.** Lin TY, Bear M, Du Z, et al. *Exp Hematol.* 2008;36:1266-1277; **3.** McCleese JK, Bear MD, Fossey SL, et al. *Int J Cancer.* 2009;125:2792-2801; **4.** Bansal H, Bansal S, Rao M, et al. *Blood.* 2010;116:4591-4599; **5.** Extra JM, Rousseau F, Bruno R, et al. *Cancer Res.* 1993;53:1037-1042; **6.** Engels FK, Ten Tije AJ, Baker SD, et al. *Clin Pharmacol Ther.* 2004;75:448-454; **7.** Baker SD, Zhao M, Lee CK, et al. *Clin Cancer Res.* 2004;10:1976-1983; **8.** Kathman SJ, Williams DH, Hodge JP, and Dar M. *Cancer Chemother Pharmacol.* 2009;63:469-476; **9.** Friberg LE, Henningsson A, Maas H, et al. *J Clin Oncol.* 2002;20:4713-4721; **10.** Taxotere® (docetaxel) injection concentrate: prescribing information. Sanofi-Aventis U.S. LLC, Bridgewater, NJ, 2010.